Abstract# 1575

CellCentric

Novel small molecule inhibitors of p300/CBP down-regulate AR and c-Myc for the treatment of castrate resistant prostate cancer

Nigel Brooks, Neil Pegg, Jenny Worthington#, Barbara Young+, Amy Prosser+, Jordan Lane+, David Taddei+, Matthew Schiewer*, Renée de Leeuw*, Jennifer McCann*, Karen Knudsen*
CellCentric Ltd, Cambridge UK; #Axis Bioservices, Coleraine, UK; +Sygnature Discovery, Nottingham UK; *The Sidney Kimmel Cancer Center at Thomas Jefferson University, Philadelphia, USA

Introduction

- Targeted degradation of androgen receptor (AR) and androgen receptor variants (ARV) remains an important therapeutic opportunity for patients with castration resistant prostate cancer.
- E1A binding protein (p300) and CREB binding protein (CBP) are two closely related histone acetyl transferase proteins that act as translational co-activators of AR.
- We have developed potent, selective and orally active small molecule inhibitors of the bromodomain of p300/CBP and report here, their impact on AR, AR-SV and c-Myc expression and function.
- We have also examined their role in driving synthetic lethality. Loss of function mutations in either p300 or CBP (present in significant proportions of lung and bladder tumours) can lead to dependency on the corresponding paralogue protein.

1. CCS series selectivity

	CCS1357	CCS1477
	In vitro probe compound	Clinical candidate
p300/CBP Kd (nM)	3.6/5.0	1.3/1.7
BRD4 Kd (nM)	245	222
Selectivity	68	170
Bromoscan @ 1μM; 32 bromodomains (% control)	BRD4 (15%); BRD1/2/3/T (15-48%) WDR (23%); SMARCA4 (7%)	BRD4 (18%); BRD1/2/3/T (15-43%) WDR (33%)
Kinome scan @10μM; 97 kinases	No significant activity	No significant activity
Cerep Safety Screen 44 @10μM	No significant activity	No significant activity

2. Inhibition of *in vitro* proliferation

Cell Line	AR status	Model	CCS1357 Proliferation IC50 μM	CCS1477 Proliferation IC50 μM
LNCaP	AR-FL	Hormone responsive	1.3	
LNCaP-AR	AR-FL over-expressed	CRPC	0.13	
VCaP	AR-FL AR-SV	CRPC	0.4	0.049
22Rv1	AR-FL AR-SV	CRPC	0.3	0.096
C42	AR-FL	CRPC	0.24	
DU145	AR negative	Hormone independent	2.7	
PC3	AR negative	Hormone independent	2.0	

Proliferation was measured with a cell viability assay (CellTiter Glo) in 22Rv1 cells (maintained in 10% FCS) after compound treatment for 72h.

3. CCS1357 degrades AR-FL & AR-SV protein & reduces expression of AR-target genes in 22Rv1 cells

a)

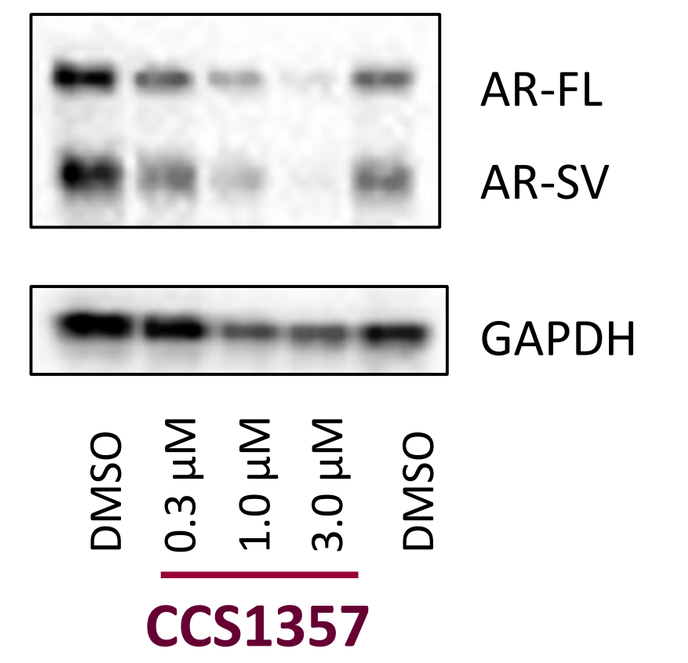
AR-FL
AR-SV
GAPDH
DMSO
0.3 μM
1.0 μM
3.0 μM
DMSO
CCS1357

b)

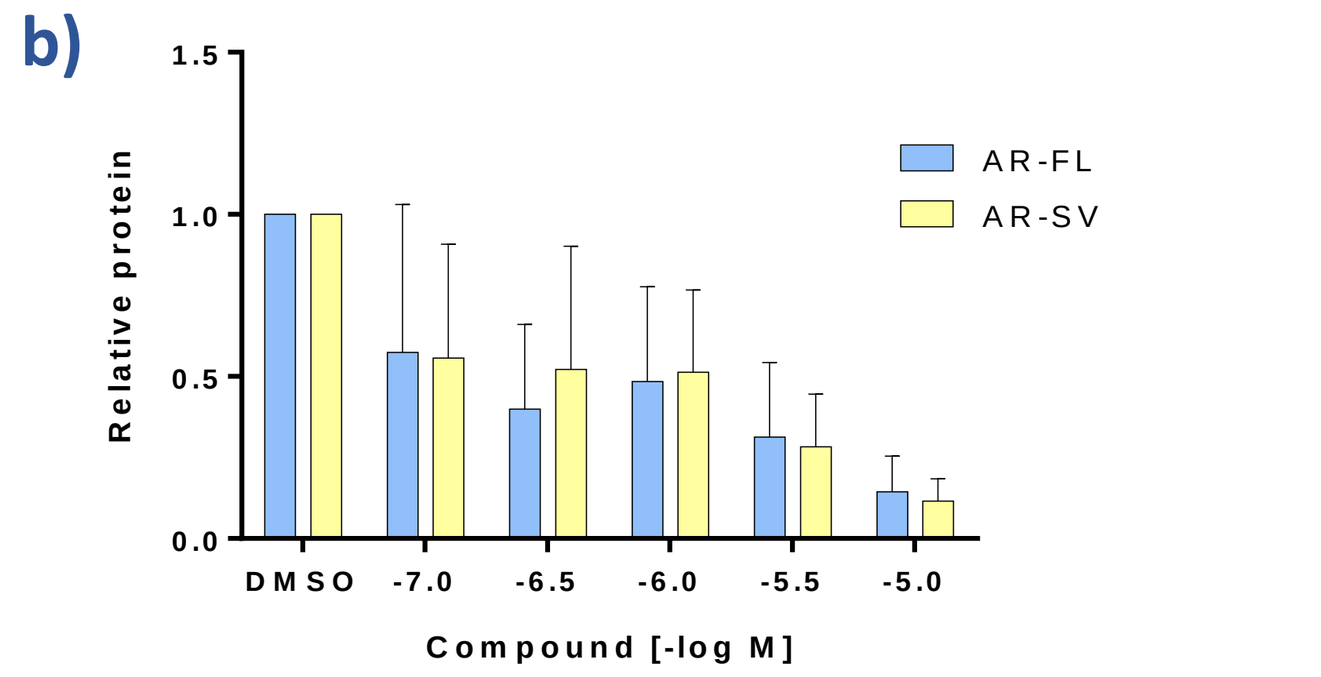
AR-FL
AR-SV
Relative protein
DMSO -7.0 -6.5 -6.0 -5.5 -5.0
Compound [-log M]

c)

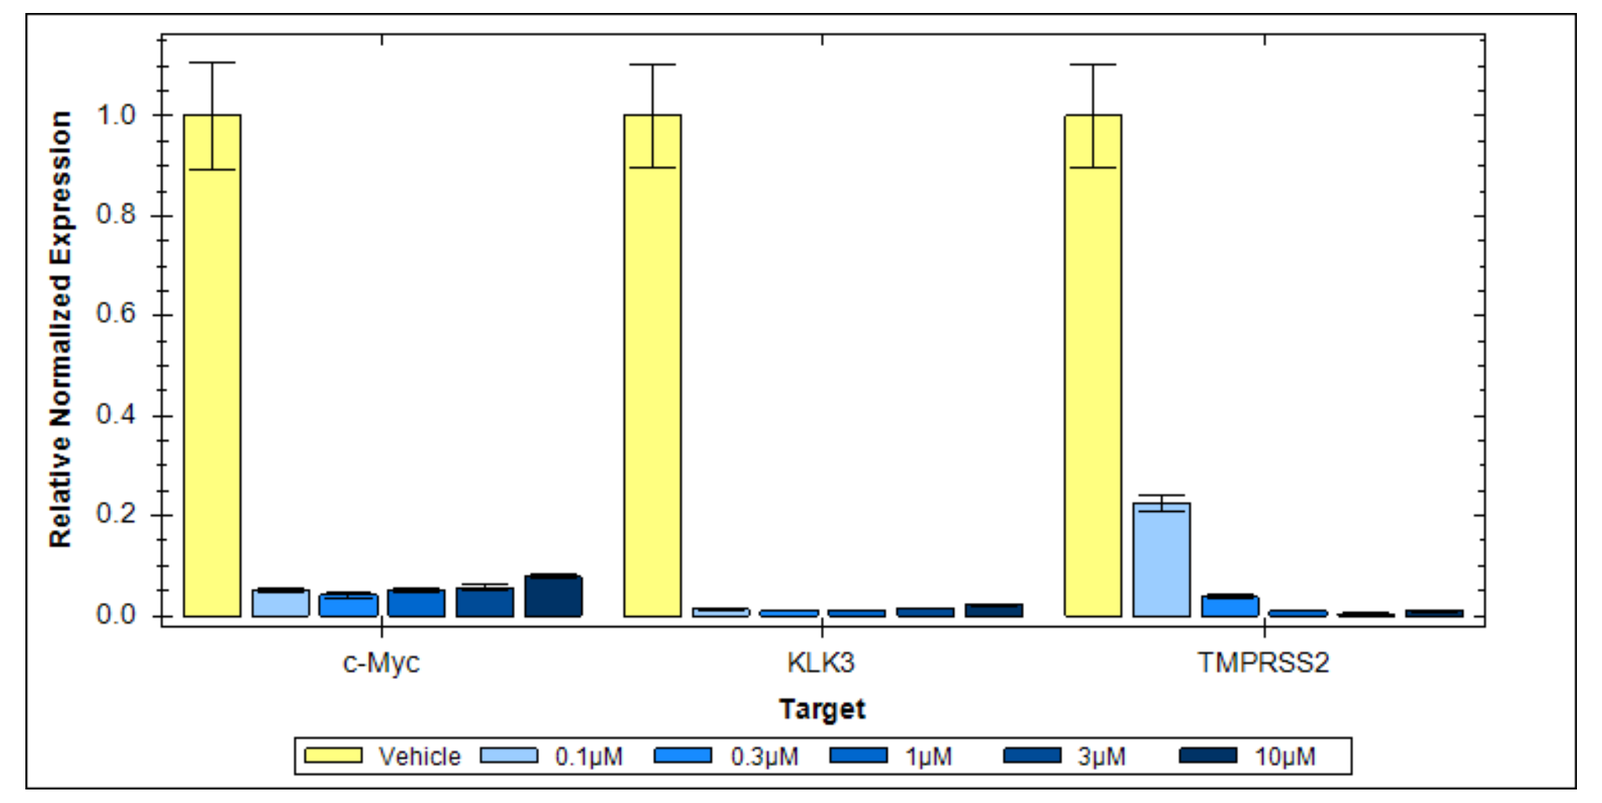
Relative Normalized Expression
c-Myc
KLK3
TMPRSS2
Target
Vehicle 0.1μM 0.3μM 1μM 3μM 10μM

a) Representative Western analysis of AR and ARSV (V7) in 22Rv1 cells after 72h of cpd treatment.
b) Ave (+/-SD) protein expression quantified by densitometry from 3 independent cell culture experiments.
c) qPCR analysis of down-stream genes after treatment with increasing doses of compound for 24h.

4. CCS1357 degrades AR & AR-SV protein; JQ1 (BRD4 inhibitor) has no effect

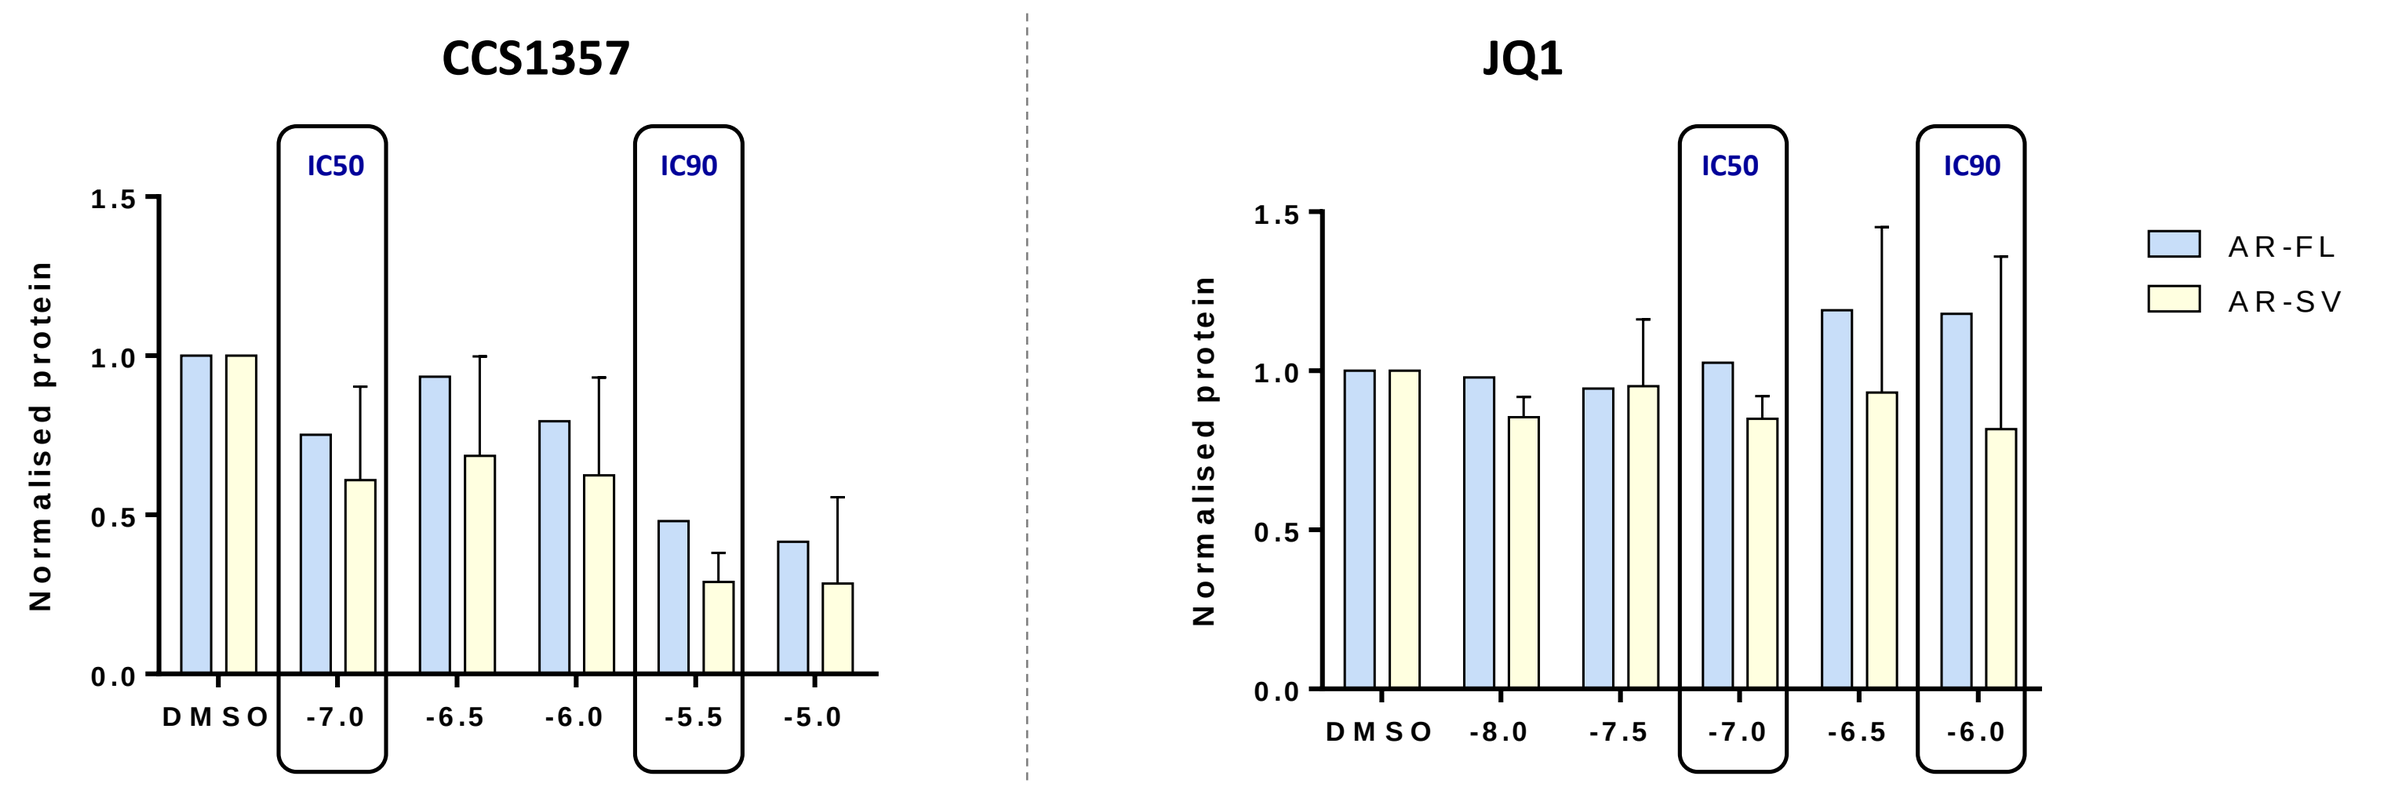
CCS1357
JQ1
IC50
IC90
Normalised protein
DMSO -7.0 -6.5 -6.0 -5.5 -5.0
DMSO -8.0 -7.5 -7.0 -6.5 -6.0
AR-FL
AR-SV

22Rv1 cells were maintained in 10% charcoal stripped FCS and treated for 72h with increasing doses of CCS1357 or JQ1. The 22Rv1 cell proliferation IC50s and IC90s for each compound are highlighted by the boxes.

5. Combination benefit of p300/CBP inhibitors with a CDK4/6 or PARP inhibitor

a)

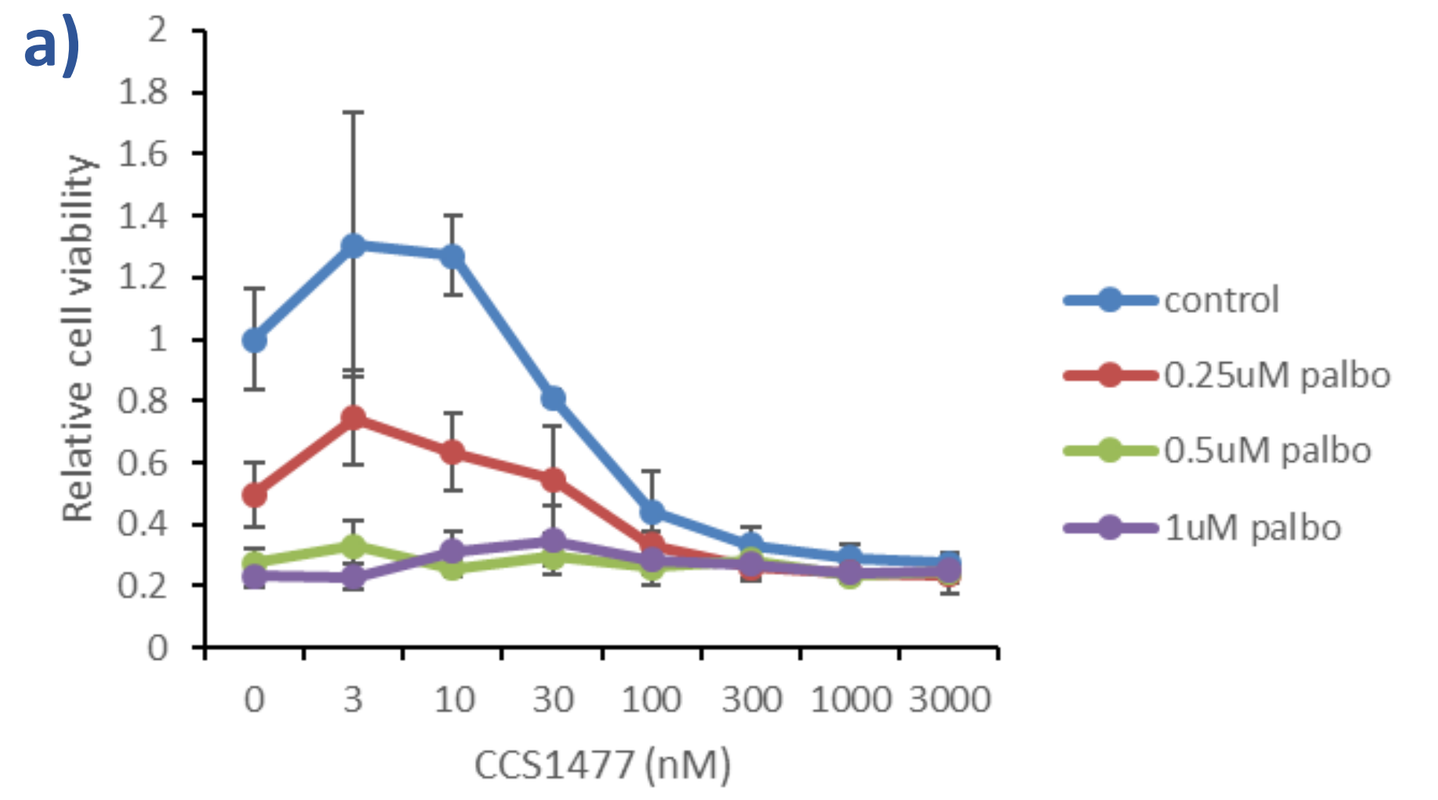
Relative cell viability
CCS1477 (nM)
control
0.25uM palbo
0.5uM palbo
1uM palbo

b)

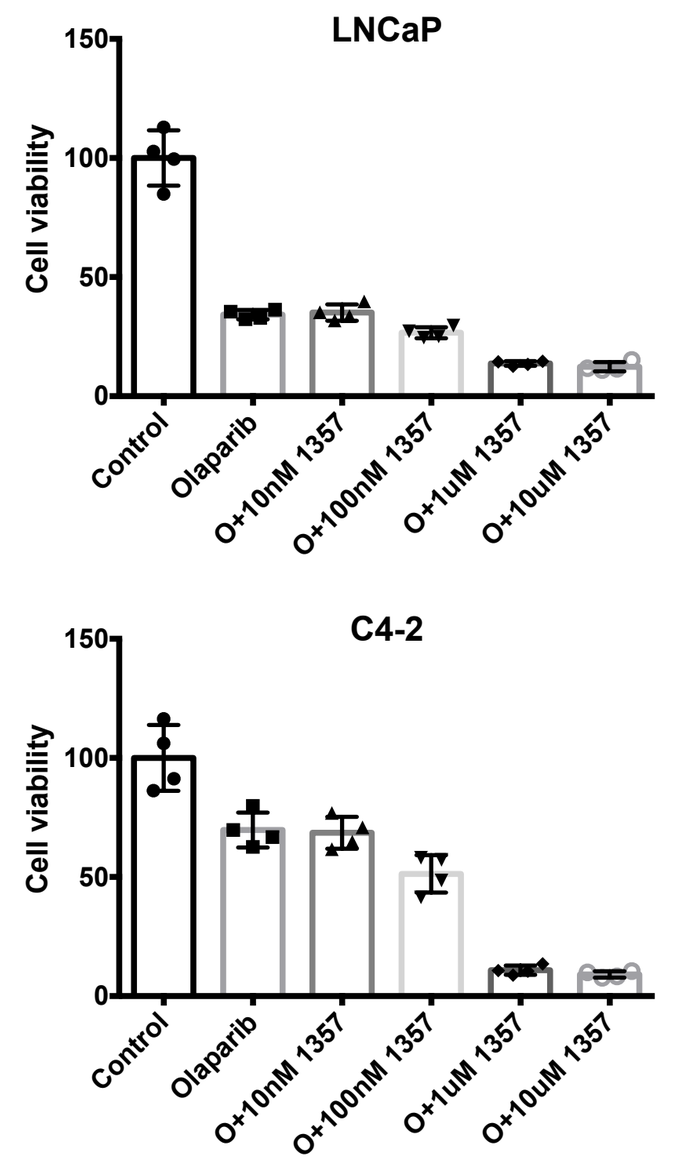
LNCaP
C4-2
Cell viability
Control
Olaparib
O+10nM 1357
O+100nM 1357
O+1uM 1357
O+10uM 1357

Cell viability was measured by picogreen assay in a) LNCaP cells treated with CCS1477 in combination with Palbociclib (CDK4/6) and in b) LNCaP or C4-2 cells treated with CCS1357 in combination with Olaparib (1uM)

6. AR, AR-SV and cMyc protein levels are reduced following single oral dose in 22Rv1 xenograft. Cleaved PARP increased at higher dose

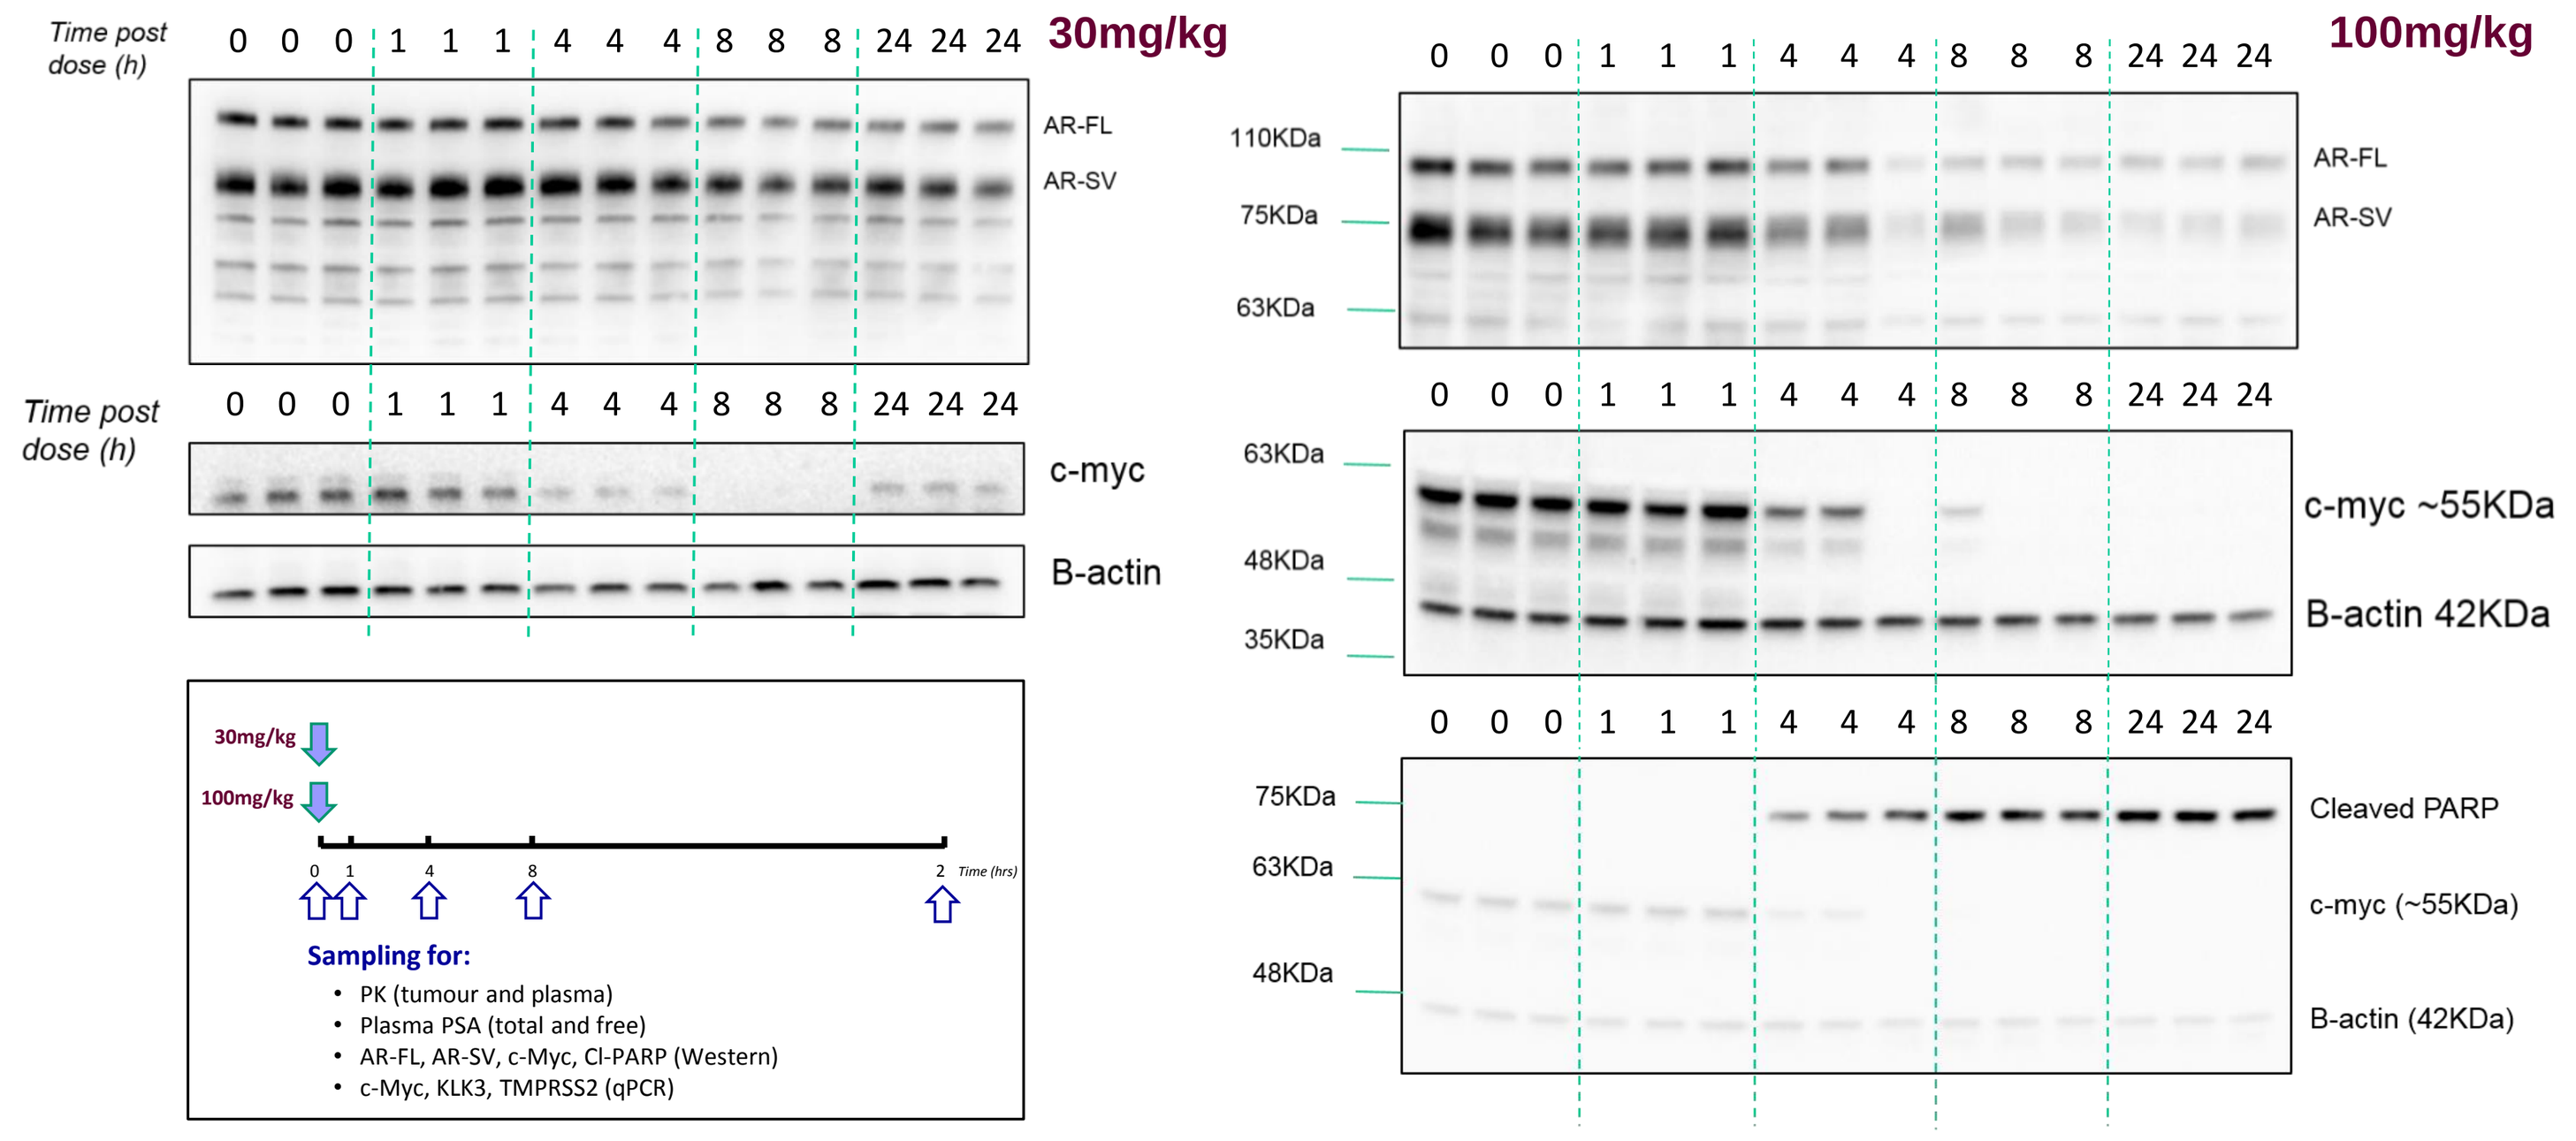
Time post dose (h)
30mg/kg
100mg/kg
AR-FL
AR-SV
c-myc
B-actin
c-myc ~55KDa
B-actin 42KDa
Cleaved PARP
c-myc (~55KDa)
B-actin (42KDa)
Sampling for:
PK (tumour and plasma)
Plasma PSA (total and free)
AR-FL, AR-SV, c-Myc, Cl-PARP (Western)
c-Myc, KLK3, TMPRSS2 (qPCR)

Protein biomarkers were measured by Western in tumour lysates collected from 22Rv1 tumours, at 1, 4, 8 and 24h after a single oral dose of CCS1477 (30 or 100mg/kg).

7. *In vivo* efficacy in 22Rv1 xenograft: Including continued tumour growth block following drug withdrawal

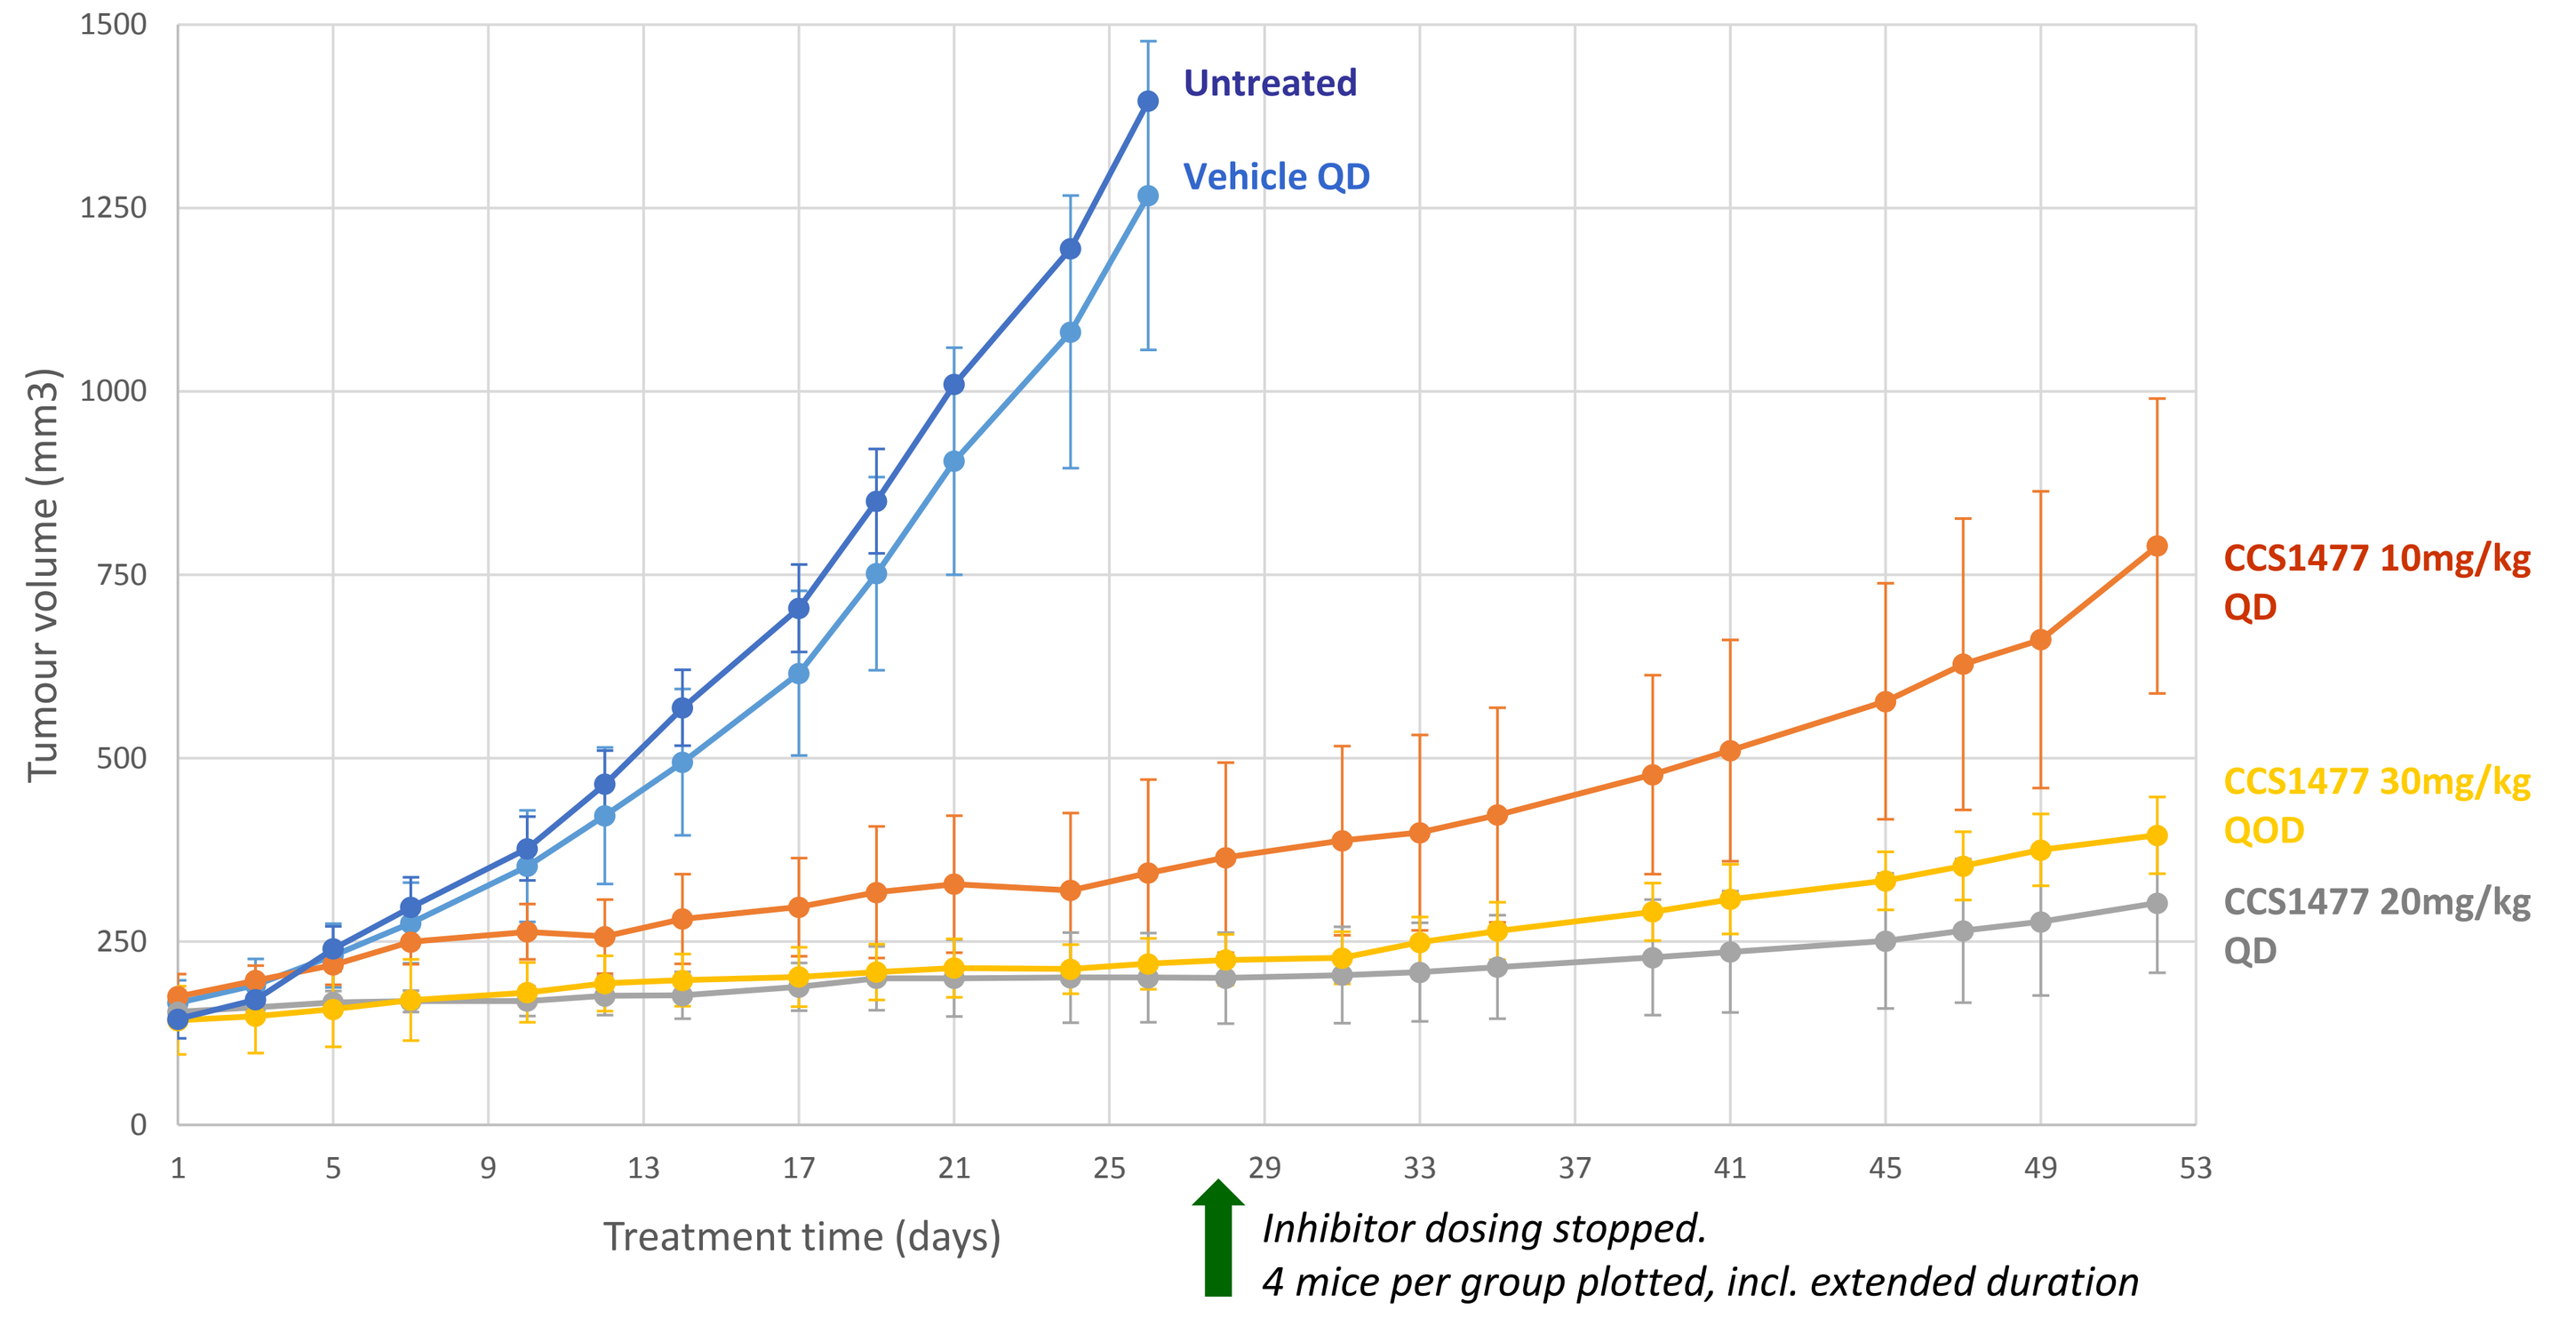
Tumour volume (mm3)
Treatment time (days)
Untreated
Vehicle QD
CCS1477 10mg/kg QD
CCS1477 30mg/kg QOD
CCS1477 20mg/kg QD
Inhibitor dosing stopped.
4 mice per group plotted, incl. extended duration

22Rv1 tumour bearing athymic nude mice were treated with CCS1477 by oral gavage, once daily (10/20mg/kg) or once every other day (30mg/kg). Vehicle (5% DMSO:95% methylcellulose [0.5%w/v]) was dosed once daily. A group of 4 untreated controls were also included.

8. Protein biomarkers are reduced in 22Rv1 tumour bearing animals treated with CCS1477 for 7 and 28 days

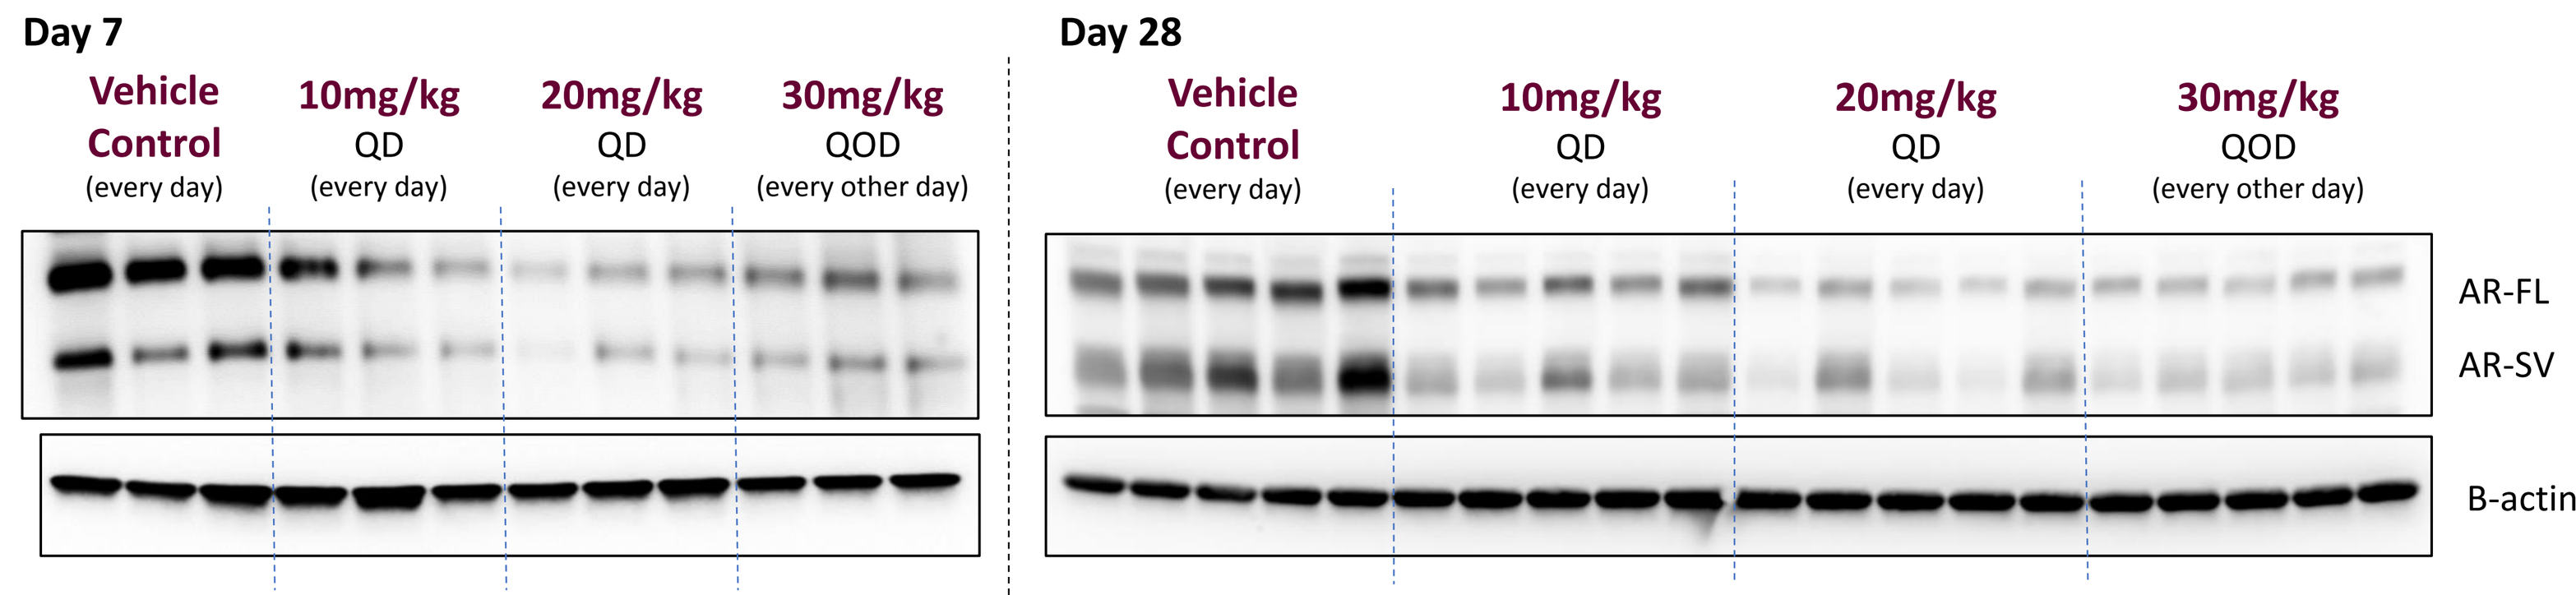
Day 7
Day 28
Vehicle Control (every day)
10mg/kg QD (every day)
20mg/kg QD (every day)
30mg/kg QOD (every other day)
AR-FL
AR-SV
B-actin

Western analysis of AR, AR-SV in 22Rv1 tumours taken from a satellite group at day 7 of the study shown in Fig. 7 and at the end of the same study at day 28.

9. Plasma PSA is reduced in 22Rv1 tumour bearing animals treated with CCS1477

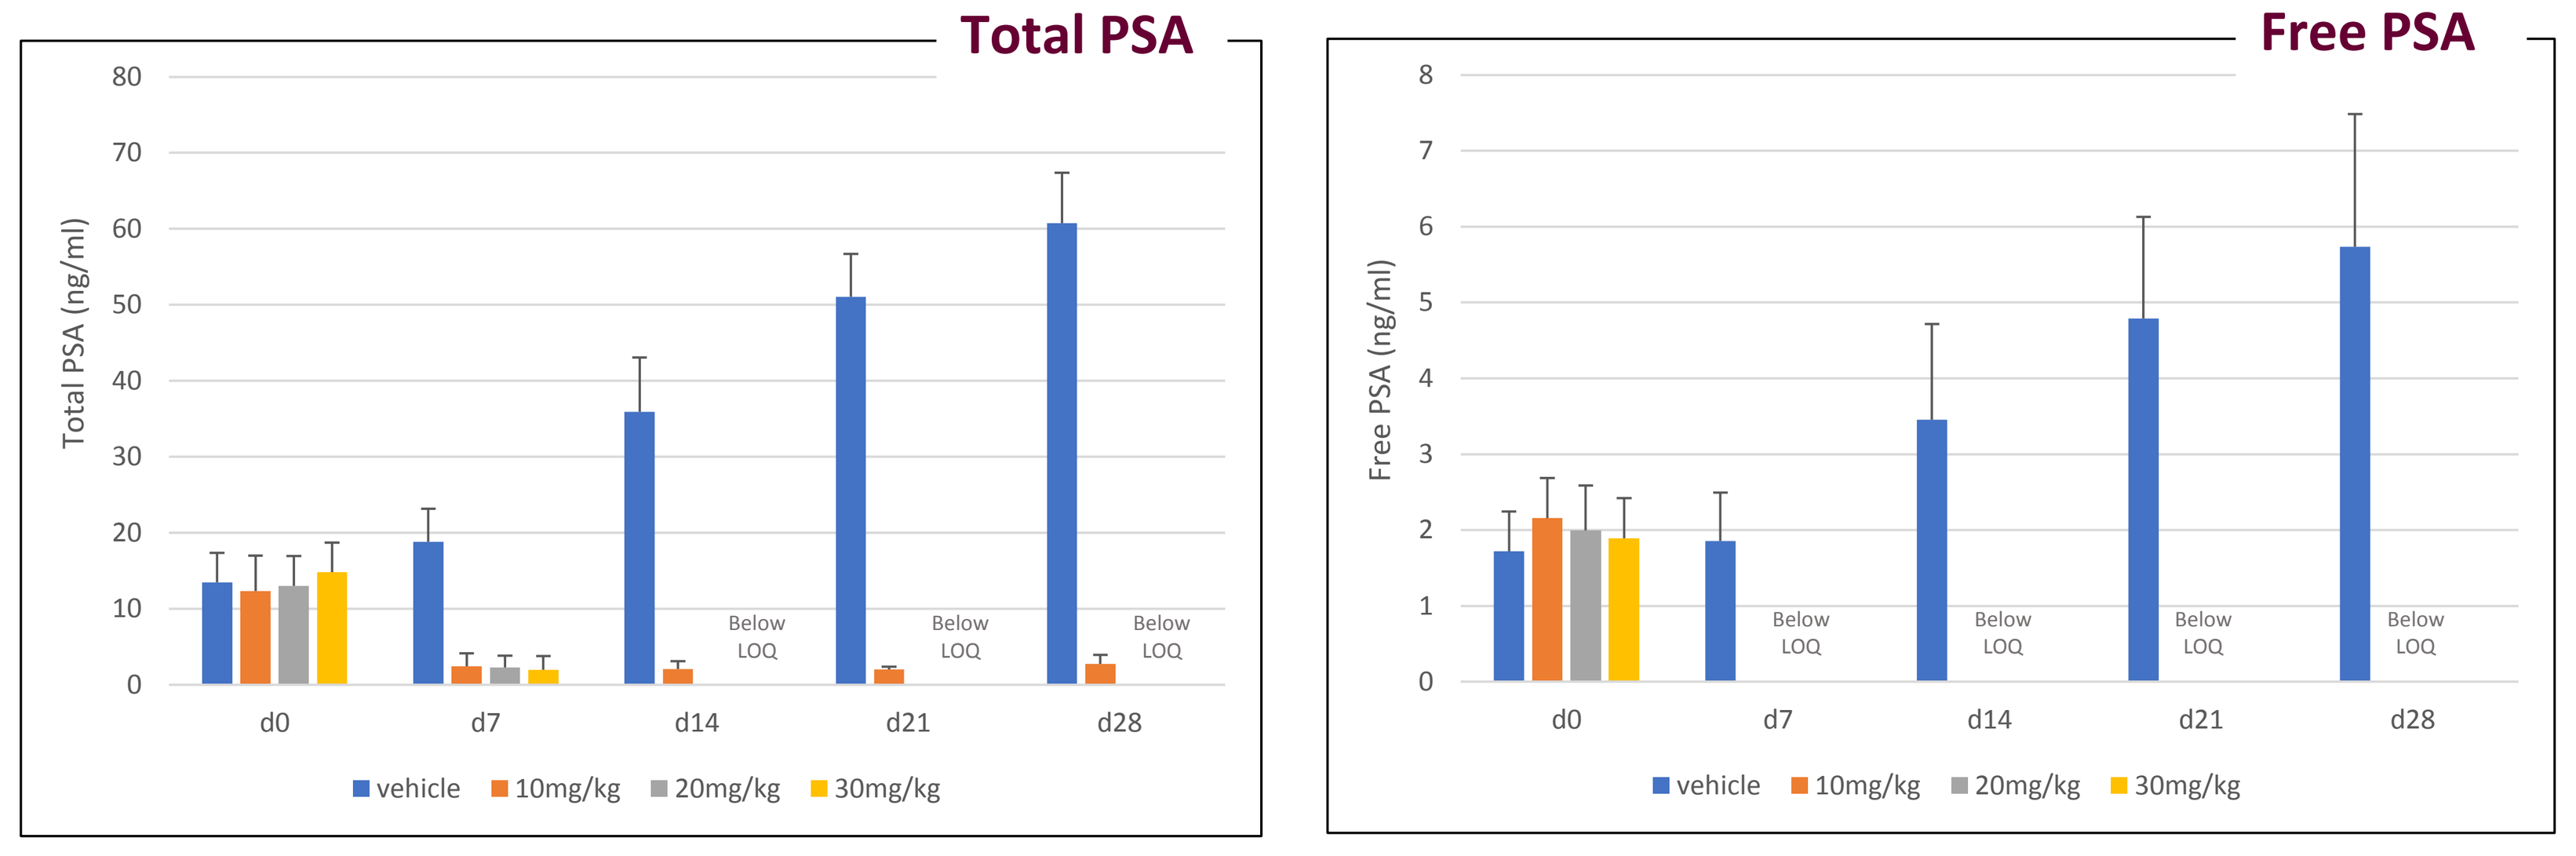
Total PSA
Free PSA
Total PSA (ng/ml)
Free PSA (ng/ml)
Below LOQ
d0 d7 d14 d21 d28
vehicle 10mg/kg 20mg/kg 30mg/kg

Plasma PSA (free and total) was measured by ELISA in blood samples collected immediately before first dosing and thereafter, at weekly intervals from the mice treated with CCS1477 in the 22Rv1 xenograft study shown in Fig. 7

10. Effects of p300/CBP and BET inhibition on cell proliferation in wild-type and CBP deficient cell lines *in vitro;* also distinct from BET inhibition (JQ1)

Cell Line	CBP mutation status	CBP30 (Proliferation IC50 (μM))	CCS1357 (Proliferation IC50 (μM))	JQ1
A549	Normal	>30	>30	0.4
H520	Deficient	>30	5.0	0.4
H1703	Deficient	>30	13.0	1.0
LK2	Deficient	3.5	0.08-0.25	0.12

Proliferation IC50 (uM) was measured with a cell viability assay (CellTiter Glo) in four lung cancer cell lines with differing CBP mutation status, after compound treatment for 72h. CBP30 = selective bromodomain inhibitor of p300/CBP from Structural Genomics Consortium (SGC).

Conclusions

- Small molecule inhibition of the bromodomain of p300/CBP, leads to down-regulation of AR, AR-SV and c-Myc, as well as inhibiting key downstream genes, including PSA and TMPRSS2.
- CCS1477, a clinical candidate, causes complete tumour growth inhibition in a 22Rv1 xenograft model at doses which are well tolerated. Inhibition is seen of tumour AR, AR-SV, as well as plasma PSA. The tumour growth inhibition caused by CCS1477 is sustained following drug withdrawal.
- AR, AR-SV and c-Myc are reduced following a single 30mg/kg dose of CCS1477. Cleaved PARP is increased at a higher dose of 100mg/kg.
- Combination benefit is observed after inhibition of p300/CBP and inhibition of either CDK4/6 or PARP.
- In lung cancer cell lines we observed differential sensitivity to CCS1357; CBP deficient lines were more sensitive (cell viability) compared with normal.
- CCS1477 is a potential first-in-class p300/CBP inhibitor for the treatment of CRPC, and potentially in the future, of tumours harbouring p300 and CBP mutations.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from AACR and the author of this poster.